P-54365

Alpha Frequency and N200 as predictors of cognitive status in a memory disorders clinic

ANNA MARIN B.A.[1,2], Kylie Schiloski B.A.[2], Naheer Lahdo B.A.[2], Ana Vives Rodriguez M.D.[2], Renee DeCaro Ph.D.[2], Andrew E. Budson M.D.[1,2], Katherine W. Turk M.D.[1,2]

[1]Department of Neurology, Boston University School of Medicine; [2]Center for Translational and Cognitive Neuroscience, VA Boston Healthcare System

Alpha peak frequency and N200 latency are useful predictors of cognitive status, especially of memory and executive function, among a memory disorders population.

INTRODUCTION

Electrophysiological measures including alpha peak power and frequency [1], and N200 latency [2] are potential biomarkers of Alzheimer's Disease (AD) pathology. It is still unclear how these measures can be used in a clinical setting to better diagnose AD and differentiate it from other pathologies. Our aim was to determine how alpha and N200 measures can help predict neuropsychological status among an heterogenous sample of patients from a memory disorders clinic.

METHODS

Data was collected using a seven-electrode COGNISION™ rig. Older veterans with memory complaints aged 50-100 were recruited from a memory disorders clinic at the VA Boston Healthcare system (N= 185) with a MMSE score of 13 and above. 28 heathy older adults were also recruited. EEG/ERP data were recorded while participants completed a three-tone auditory oddball task. Participants who failed an audiometry screening test were excluded. All subjects were administered a neuropsychological battery to assess degree of cognitive impairment. Amyloid PET (aPET) results were also available for a subset of subjects (N=67). 26 subjects had an abnormal aPET scan, consistent with a diagnosis of AD.

N=213	Age	Years of Education	MMSE	Button Press Accuracy (%)	False Alarms (%)	Median RT (*ms*)	GDS	GAI
Mean (SD)	73.1 (8.1)	14.3 (2.8)	25.0 (3.6)	89.7 (17.5)	2.4 (6.5)	502.6 (129.4)	4.3 (3.5)	5.2 (5.8)

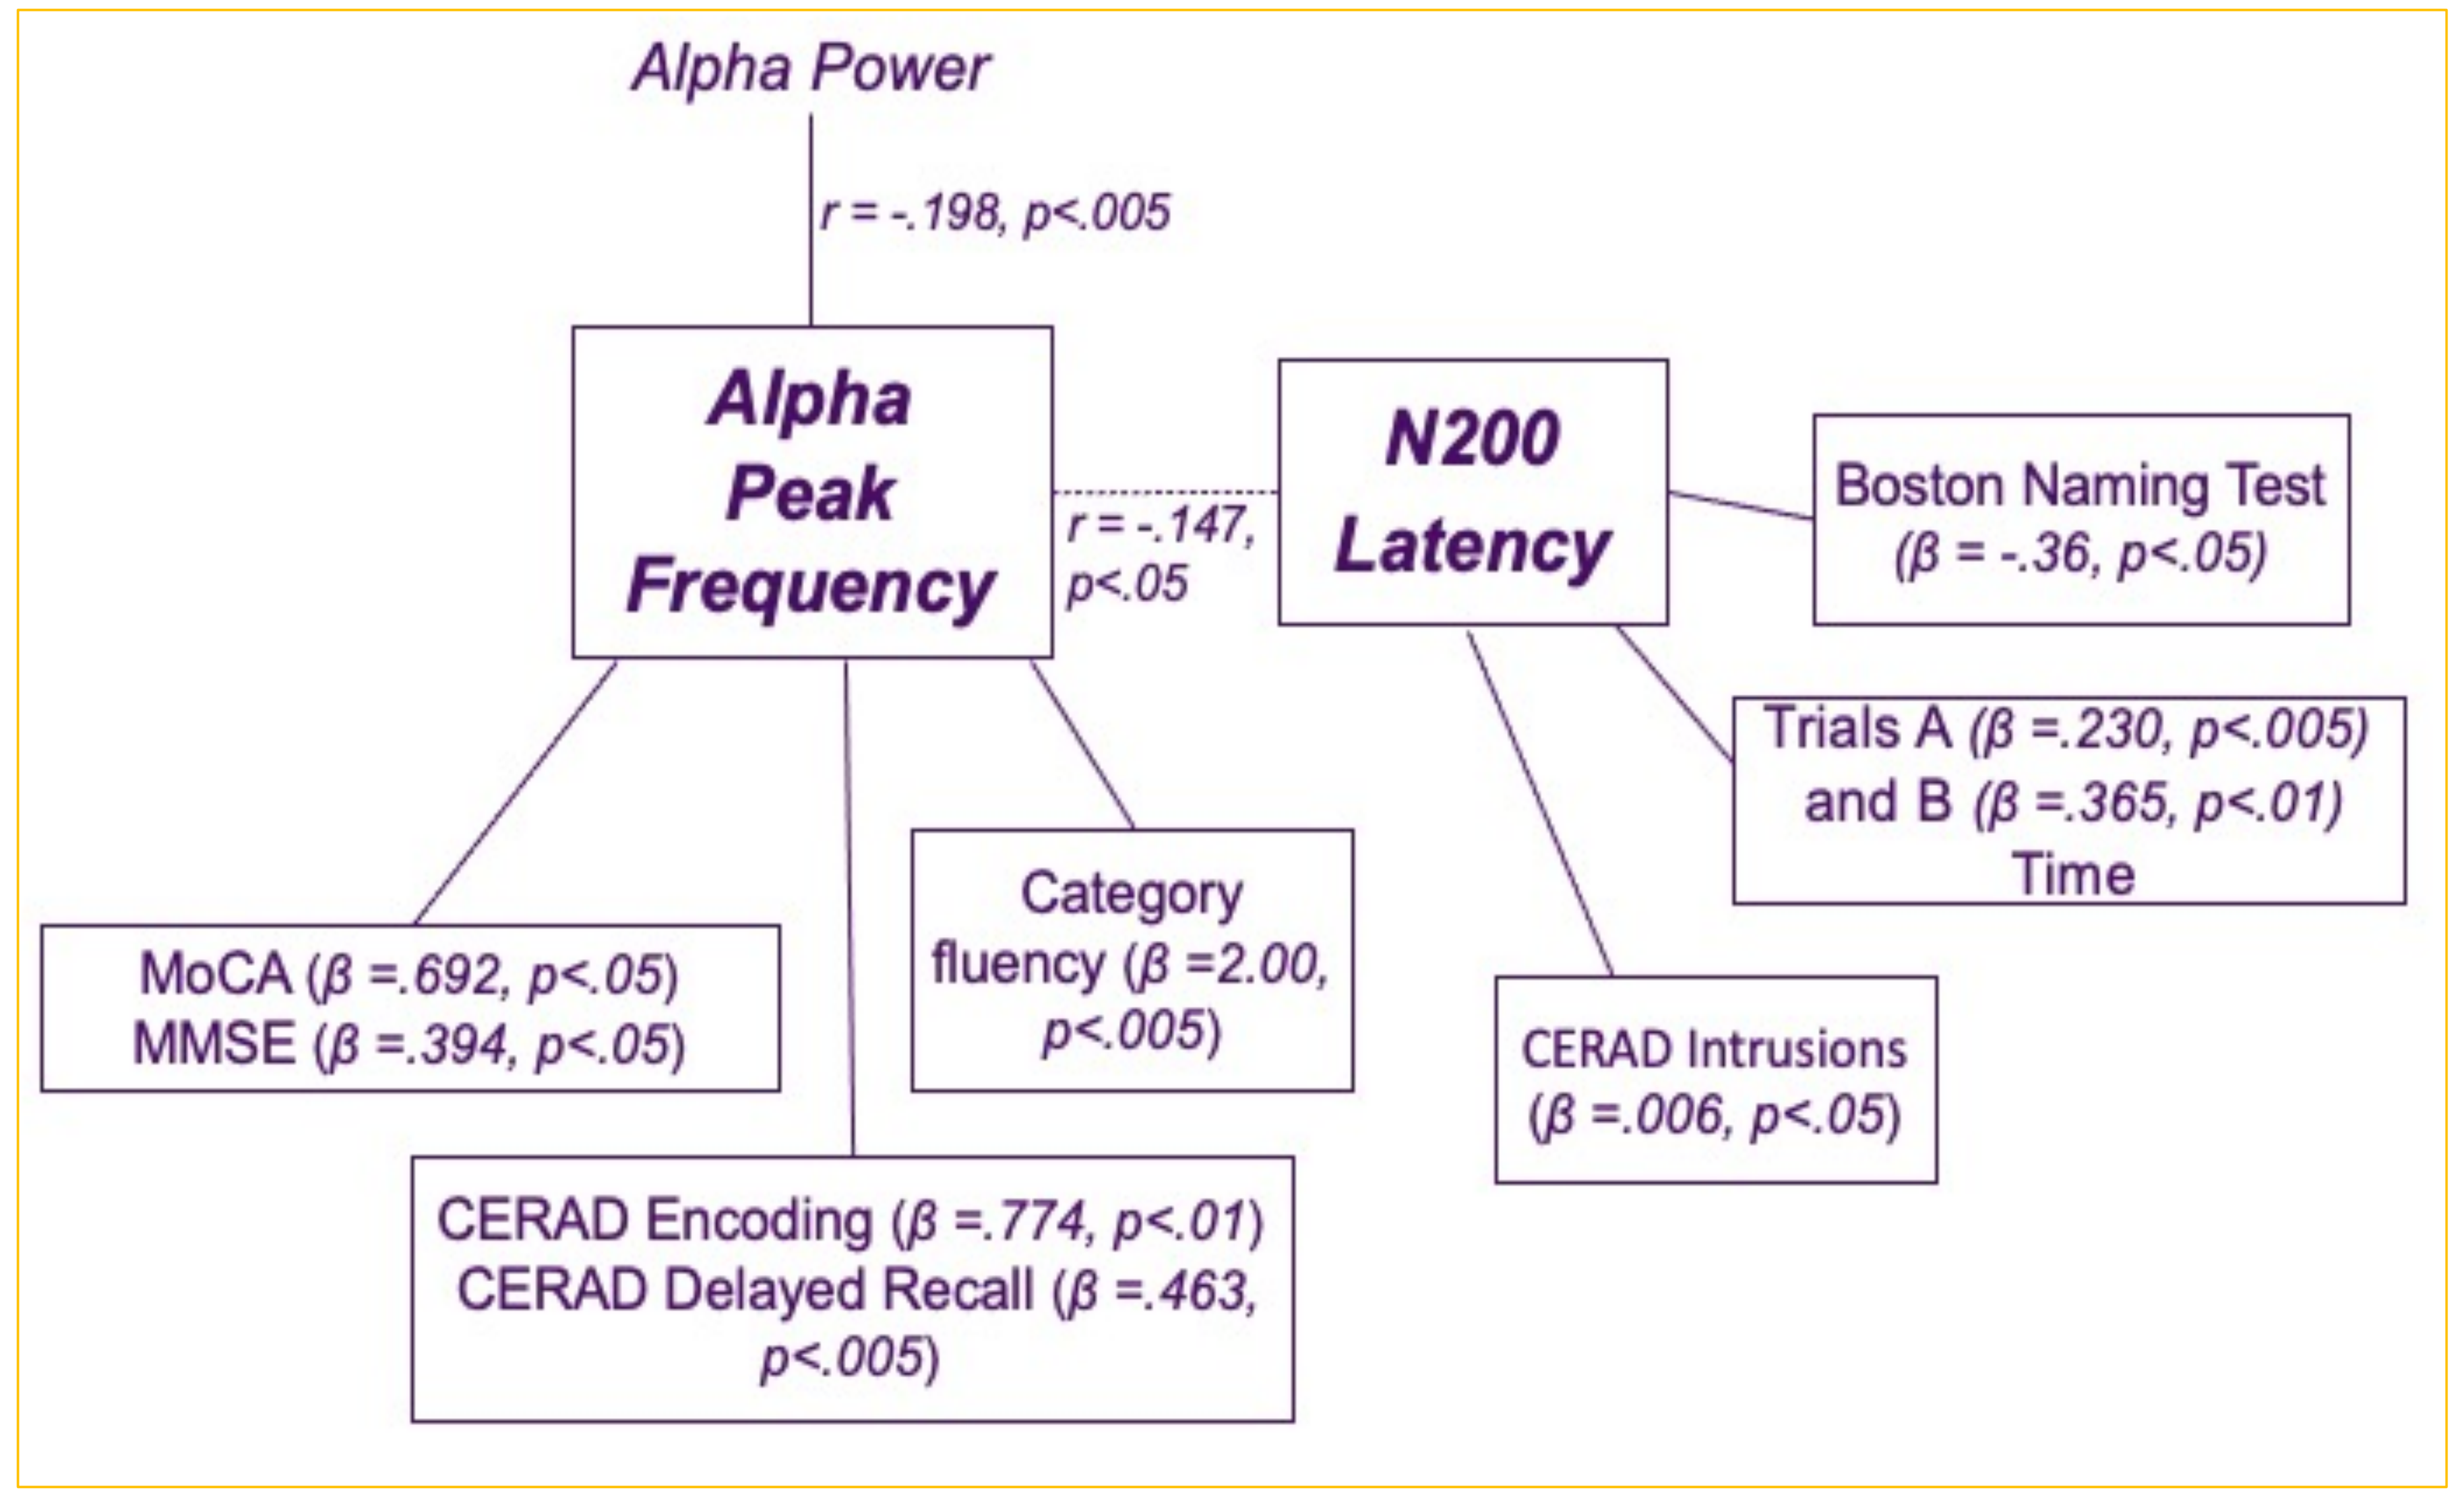
Alpha Power
r = -.198, p<.005
Alpha Peak Frequency
r = -.147, p<.05
N200 Latency
Boston Naming Test (β = -.36, p<.05)
Trials A (β =.230, p<.005) and B (β =.365, p<.01) Time
CERAD Intrusions (β =.006, p<.05)
MoCA (β =.692, p<.05)
MMSE (β =.394, p<.05)
Category fluency (β =2.00, p<.005)
CERAD Encoding (β =.774, p<.01)
CERAD Delayed Recall (β =.463, p<.005)

KEY FINDINGS

Linear regressions corrected for age and education were computed for all cognitive measures that correlated with alpha peak frequency and N200 latency. Results are outlined in the figure to the left. Results also indicated a non-significant trend for alpha peak frequency as a predictor of aPET status ($\chi2(3)=5.02, p=.083$) using a binary logistic regression.

CONCLUSIONS

Abnormalities in alpha peak frequency and N200 latency EEG signals are useful predictors of cognitive impairment, especially in the domains of memory and executive function. Overall, these neural measures represent a novel tool to better assess memory disorders patients,and characterize the cognitive profile of their underlying pathology in a heterogenous clinical setting.

REFERENCES

[1] Dierks, T., Ihl, R., Frölich, L., & Maurer, K. (1993). Dementia of the alzheimer type: Effects on the spontaneous EEG described by dipole sources. *Psychiatry Research: Neuroimaging*, *50*(3), 151–162. https://doi.org/10.1016/0925-4927(93)90027-F

[2] Cecchi, M., Moore, D. K., Sadowsky, C. H., Solomon, P. R., Doraiswamy, P. M., Smith, C. D., Jicha, G. A., Budson, A. E., Arnold, S. E., & Fadem, K. C. (2015). A clinical trial to validate event-related potential markers of Alzheimer's disease in outpatient settings. *Alzheimer's & Dementia : Diagnosis, Assessment & Disease Monitoring*, *1*(4), 387–394.https://doi.org/10.1016/j.dadm.2015.08.004

ACKNOWLEDGEMENTS

Funded by Cyclerion Therapeutics, Inc., and the Alzheimer's Association.

CONTACT

Anna Marin

alzheimer's association®

AAIC 21 ALZHEIMER'S ASSOCIATION INTERNATIONAL CONFERENCE®